TREATING OCULAR GRAFT VERSUS HOST DISEASE IN AN 8-YEAR-OLD PATIENT WITH SCLERAL LENSES

École d'optométrie
Université de Montréal

Gabriella Courey; Langis Michaud, OD MSc FAAO (Dipl)

École d'optométrie, Université de Montréal

Introduction

Patients with graft versus host disease (GvHD) are strongly inclined to develop ocular surface disease, particularly dry eye. (1) In refractive cases where conventional dry eye therapy is ineffective, treatment of the ocular surface includes scleral lenses. (2) The following case report highlights the use of scleral lenses to treat ocular GvHD and manage dry eye symptoms in an 8-year-old patient.

Subjective

L.L., an 8-year- old female, suffers from ocular GvHD secondary to three recurring cases of leukemia and presented signs and symptoms of severe Dry Eye Syndrome (DES). The patient was extremely symptomatic of her DES despite her constant use of non-preserved artificial tears (carboxylmethylcellulose, 1.4%, Allergan, Ireland; dosage q1h00), topical compound albumin tears derived from serum (6-8 times/day), topical cyclosporine (Restasis 0,05%, dosage BID) and petroleum gel (Systane Ointment, Alcon, TX, dosage qhs), so much so that she had to keep her eyes closed for the majority of waking hours. Her systemic medication consisted of prednisone 20 mg/day.

Objective

Entering uncorrected visual acuity (VA) was OD: 20/30 and OS: 20/40 using the Snellen VA chart under photopic illumination, not improvable with refraction and similar using pinhole. Fundus examination was unremarkable.

+ *References.*

1. Bradfield YS, Kushner BJ, Gangnon RE. Ocular complications after organ and bone marrow transplantation in children. J AAPOS. 2005;9:426–432.
2. Courey C, Michaud L. (2017). Variation of clearance considering viscosity of the solution used inthe reservoir and following scleral lens wear over time. Contact Lens and Anterior Eye.
3. Tangible Hydra-PEG Technology. URL : www.tangiblescience.com/copy-of-technology-1.(Retrieved on Dec 12 2017).

FIGURE 1 – Pentacam Holladay Report scans for OD (A) and OS (B). Scanning was limited at the initial visit due to the fact that the patient's eyes were closed the majority of the time. Scans show extremely irregular corneas due to severe tear film insufficiency.

(A)

(B)

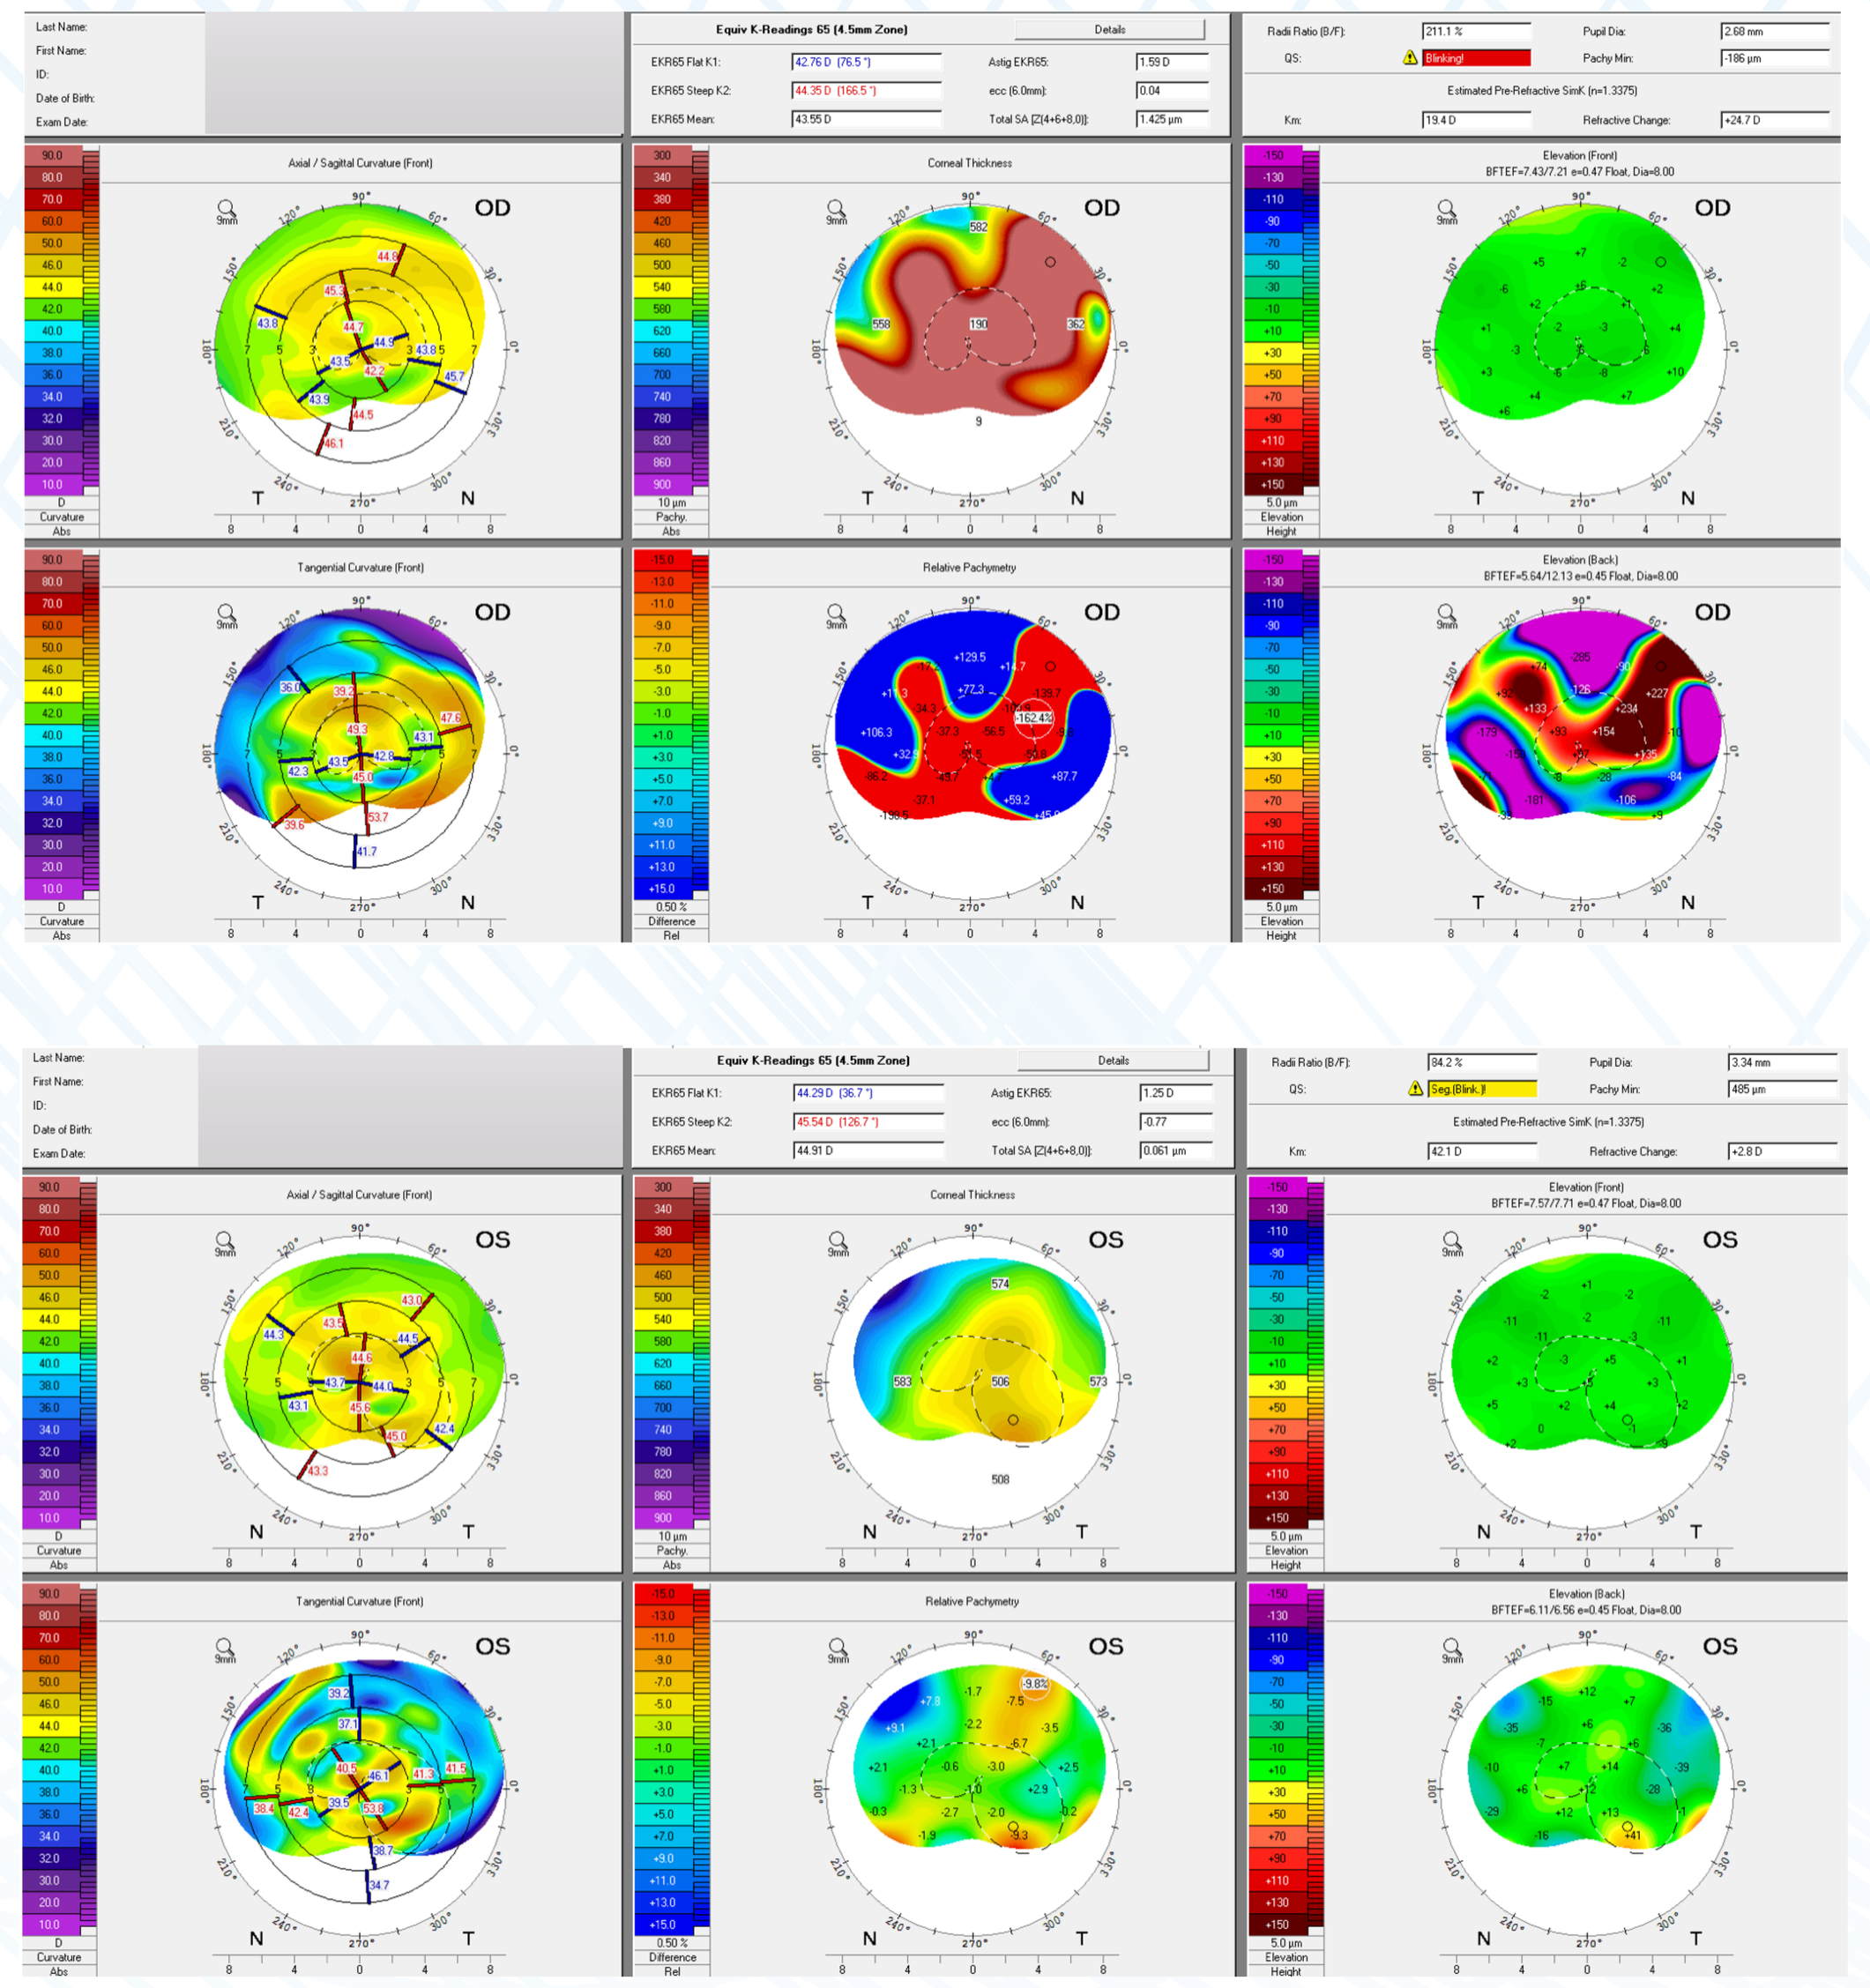

IMAGE 1 – Anterior segment image depicting severe filamentary keratitis

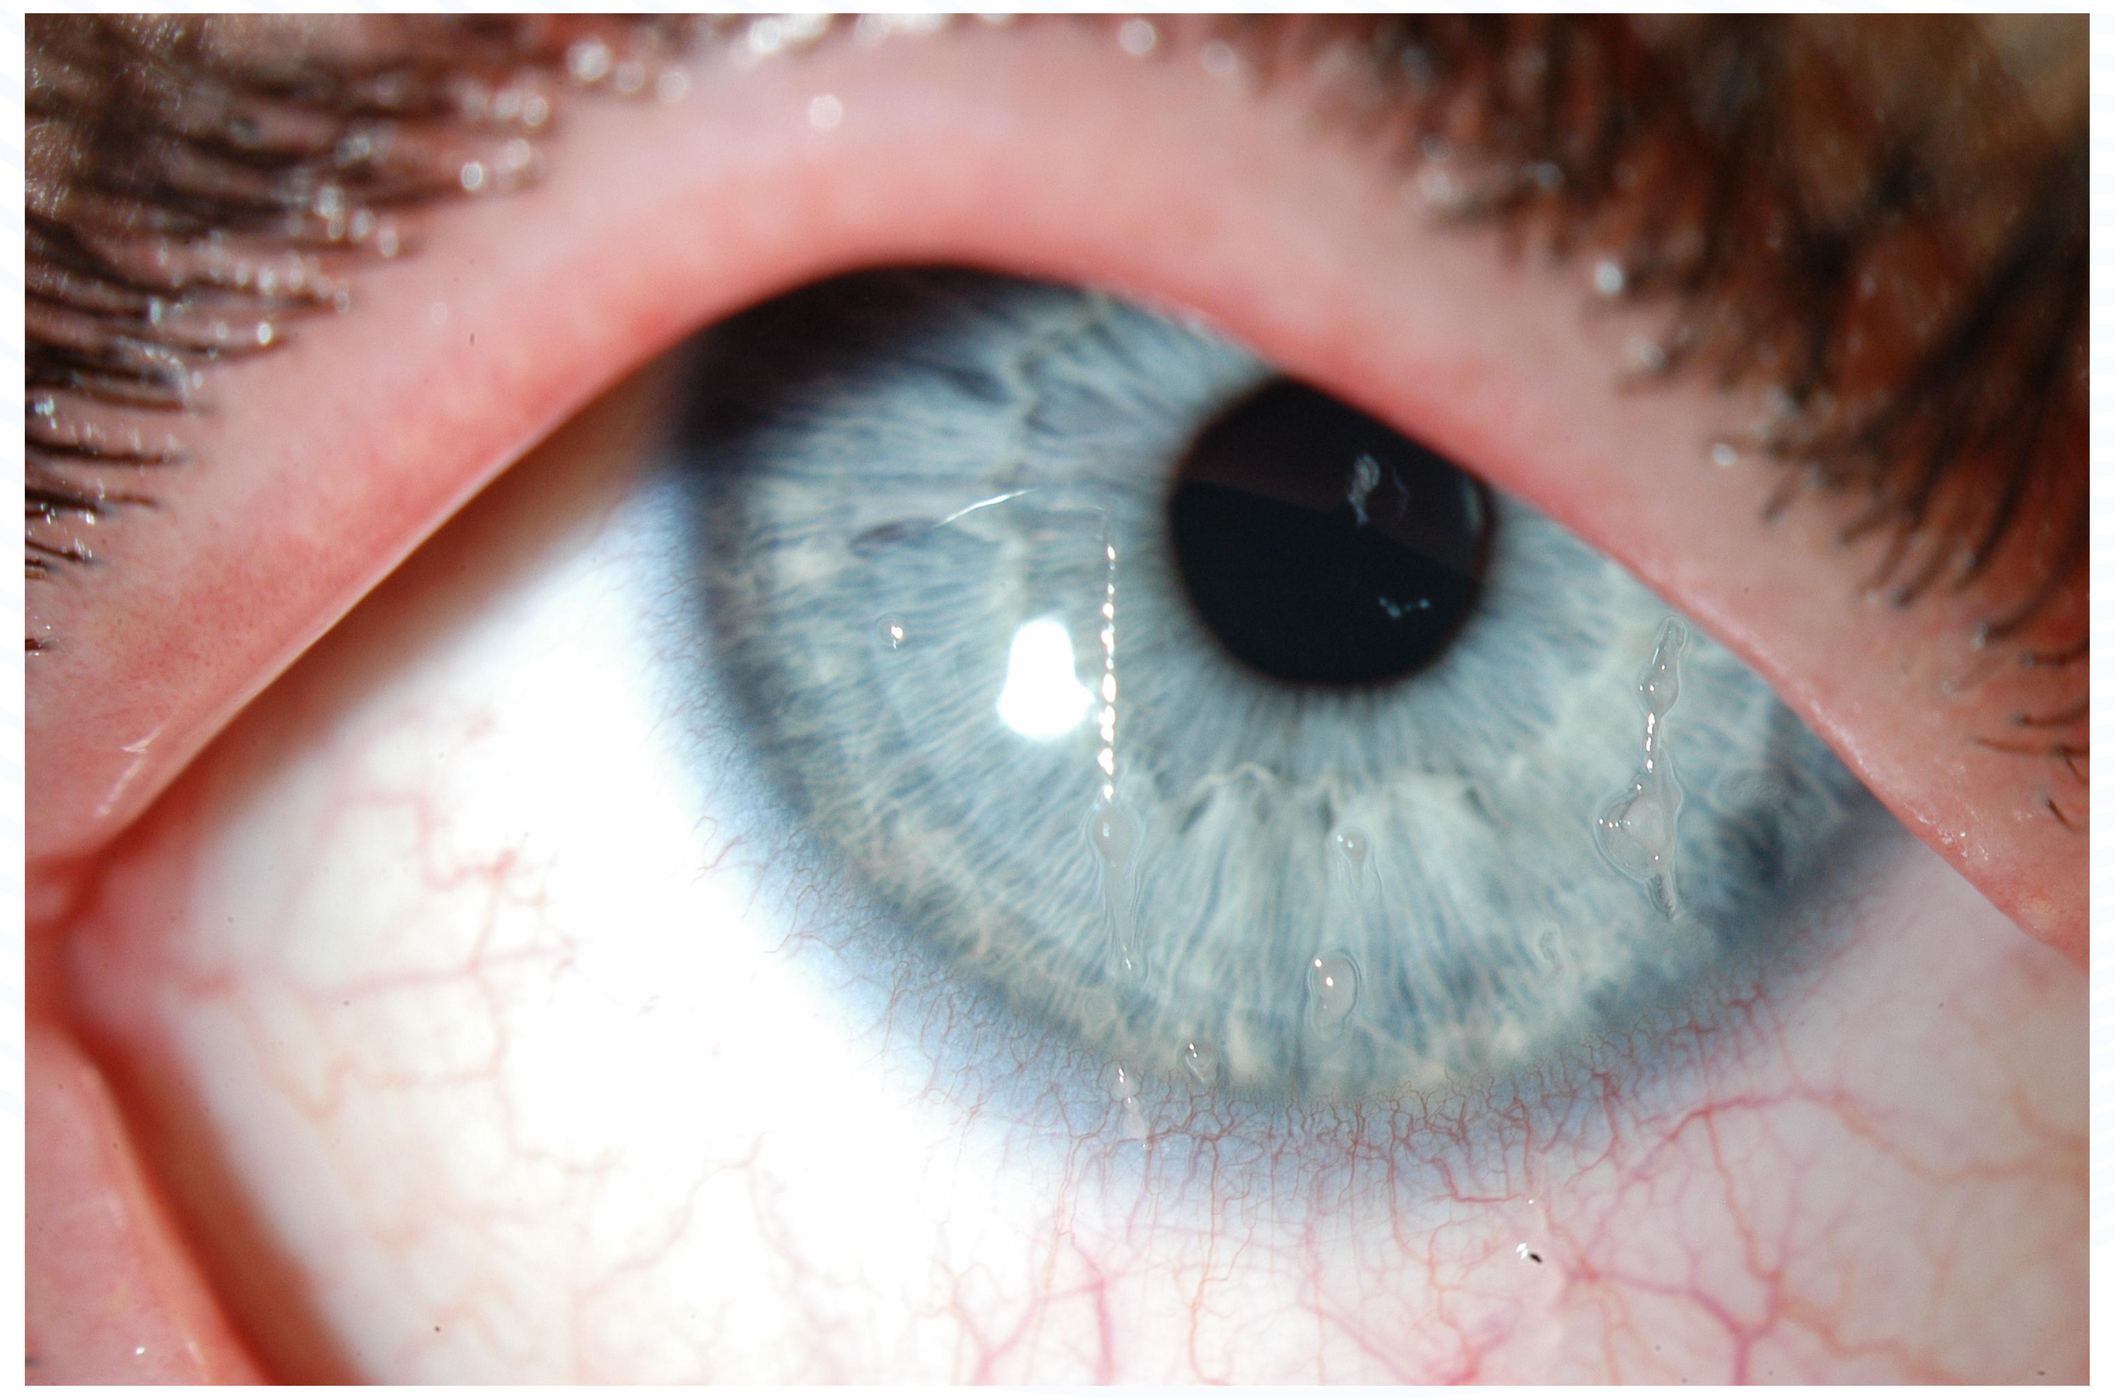

Assessment

This patient presents a filamentary keratitis secondary to chronic eye dryness, related to her systemic condition (GvHD).

Treatment Plan

Larger scleral contact lenses (> 16 mm) are considered as part of the DES treatment, once lubrication and medication are not sufficient in reducing the signs and the symptoms felt by the patient. Considering the condition of the patient and the reduced lid aperture, a smaller lens option was considered.

Scleral Lens Trials

+ OneFit 2.0 mini-scleral lens (Blanchard Laboratories, Sherbrooke, Quebec)
+ The trial lenses were selected based on the manufacturer's recommendations: 0.2 mm flatter than the central flat K. After lens insertion, the patient was encouraged to keep her eyes open and lenses were evaluated after 30 minutes of wear (see Table 1).

TABLE 1 – INITIAL LENS SELECTION

	OD	OS
BASE CURVE	7.8 mm	7.6 mm
DIAMETER	14.9 mm	14.9 mm
LENS POWER	-2.50 D	-3.00 D
MATERIAL	Boston XO	Boston XO
OVER-REFRACTION	+3.75 (20/25)	+2.75 (20/25)
OCT ANTERIOR SEGMENT (CLEARANCE)	166 µm	241 µm

+ Clearance over the right eye was deemed insufficient; thus, the base curve was steepened by 0.1 mm and the lens power was compensated according to the manufacturer's guidelines (see Table 2).

TABLE 2 – FINAL LENSES

	TYPE OF LENS	BASE CURVE	DIAMETER	LENS POWER
OD	OneFit 2.0	7.7 mm	14.9 mm	-0.75 D
OS	OneFit 2.0	7.6 mm	14.9 mm	+0.25 D

+ The lenses were ordered with Hydra-PEG coating considering the amount of filaments and other deposits generated by her eyes. This treatment improves wettability, increases lens surface water retention, lubricity and minimizes deposits and lens fogging. (3)
+ At dispensing, the lens was evaluated 30 minutes post-insertion. The patient noted extreme comfort and was pleasantly surprised that she could finally keep her eyes open 30 minutes after insertion. Her distance monocular and binocular VA's with the lenses were 20/20.
+ Lens maintenance consisted of soaking the lenses every night overnight in hydrogen peroxide solution. Before insertion, L.L. was instructed to rinse the lens with non-preserved saline solution and proceed to fill it with drops of sodium-hyaluronate based non-preserved artificial tears (Hylo-Dual, Candor Vision), which was also recommended to facilitate lens removal. In conjunction with the scleral lenses, the patient was advised to continue using Restasis BID while only using albumin before bedtime. Follow-up was scheduled in 4 weeks' time.

FIGURE 2 - Optovue RTVue OCT images of central clearance over OD (A) and OS (B) 4 hours post insertion at follow-up visit.

(A)

(B)

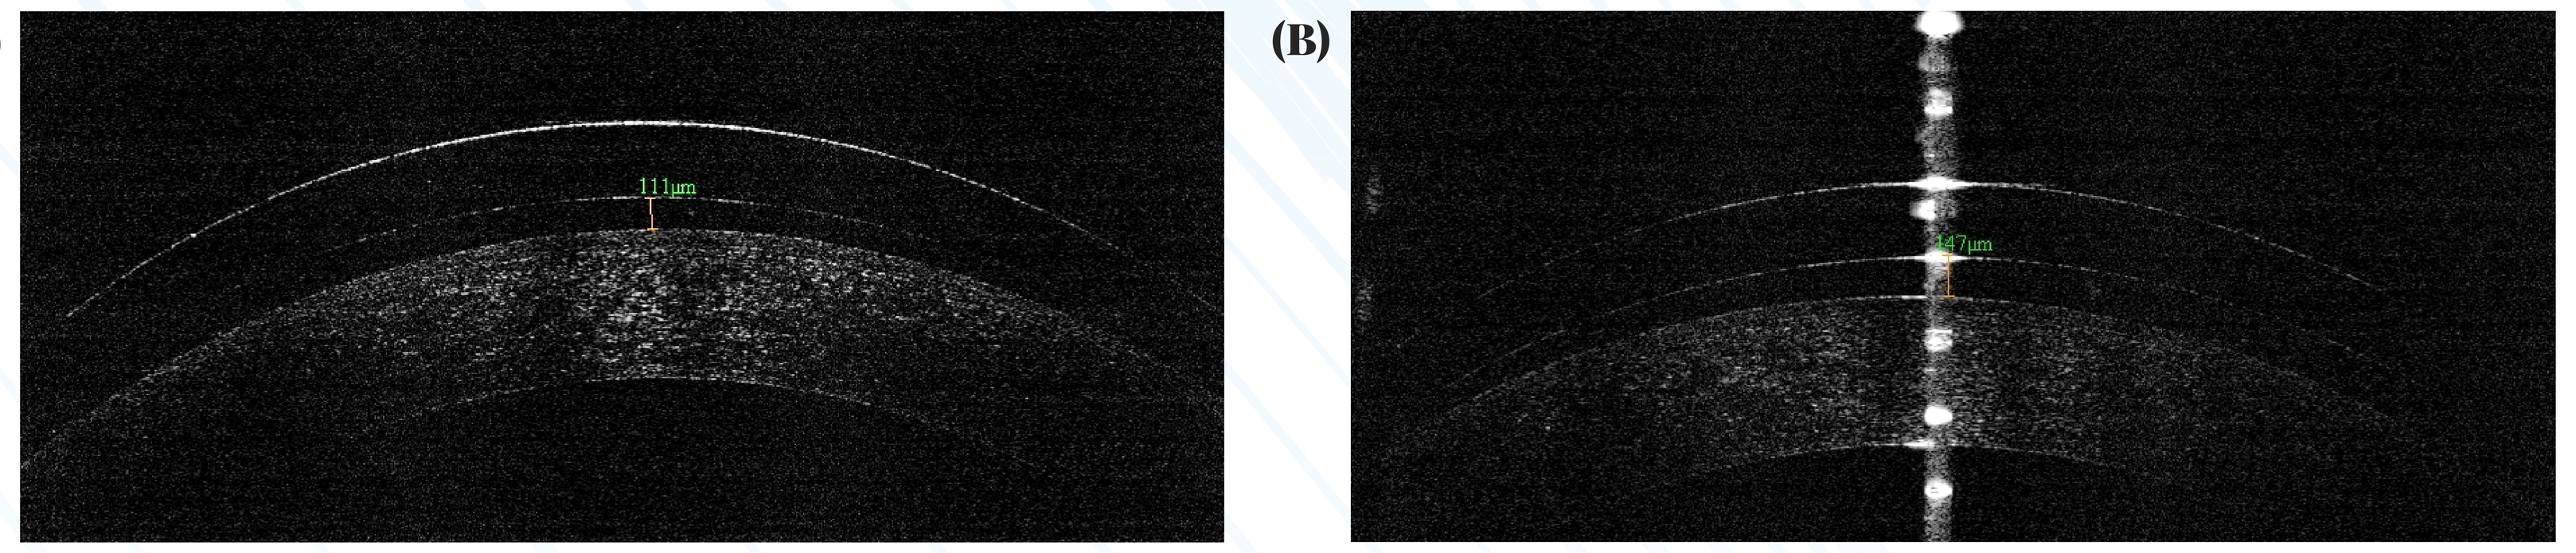
111µm
147µm

Follow-Up

+ The patient maintained monocular and binocular VA's of 20/20 while happily noting that she was extremely comfortable and could keep her eyes open a whole day without DES symptoms. She also proudly stated that she manipulated the lenses independently and wears them 10-12 hours/day without any signs of ocular dryness, fluctuating vision, redness, pain or discomfort during her 4 weeks of wear.
+ After 4 hours of wear, ocular examination revealed that the lens was centered without compression on either the nasal or temporal bulbar conjunctiva. The central clearance was evaluated at 111 µm OD and 147 µm OS (see Figure 2).
+ The need for artificial tear lubrication dropped significantly, which, alone, improved the quality of life of the patient and her entire family.

Discussion & Conclusion

As demonstrated by this patient's successful outcome, scleral lenses have proven themselves as an effective tool to manage patients with severe ocular dryness. For this patient, the improvements in both VA and ocular comfort provided by the mini-scleral lens changed L.L.'s quality of life. From barely being able to keep her eyes open for a couple of seconds in a row, these lenses allowed her eyes to remain open without the constant application of ocular lubricants, all factors which play a significant role in giving L.L. more opportunities in the future. For similar cases, a thorough case history and observation of clinical signs will help practitioners choose a scleral lens that is right for the patient after all other options have been explored.

GLOBAL Specialty Lens Symposium presented by Contact Lens SPECTRUM